Efficacy and Safety of Once-Daily Opicapone in Older Patients with Parkinson's Disease and Motor Fluctuations

Jacquelyn Bainbridge,[1] Stephen R. Saklad,[2] Olga Klepitskaya,[3] Eric Jen,[3] Fabrizio Stocchi[4]

[1]Department of Clinical Pharmacy, University of Colorado School of Medicine, Aurora, CO; [2]College of Pharmacy, University of Texas at Austin, San Antonio, TX; [3]Neurocrine Biosciences, Inc., San Diego, CA; [4]Department of Neurology, IRCCS San Raffaele Pisana, Rome, Italy

ABSTRACT DESCRIPTION

Pooled data from 2 pivotal Phase 3 studies of opicapone in patients with Parkinson's disease (NCT01568073, NCT01227655) were analyzed to evaluate efficacy and safety of opicapone in older participants (≥65 years). Older participants had significantly greater improvements in "OFF"-time and "ON"-time with once-daily opicapone versus placebo; opicapone was generally well-tolerated.

INTRODUCTION

- Levodopa is considered the most effective treatment for managing Parkinson's disease (PD), yet patients still often experience motor fluctuations as the disease progresses[1]
- Opicapone (ONGENTYS®) is a once-daily catechol-O-methyltransferase (COMT) inhibitor approved in the United States as adjunctive treatment to levodopa/carbidopa in patients with PD experiencing "OFF" episodes[2]
- The efficacy, safety, and tolerability of once-daily opicapone in patients with PD and motor fluctuations on a stable regimen of levodopa/dopa decarboxylase inhibitor were demonstrated in 2 double-blind, placebo-controlled, pivotal Phase 3 clinical trials, BIPARK-1 (NCT01568073)[3] and BIPARK-2 (NCT01227655)[4]
- This post-hoc analysis evaluates the efficacy and safety of adjunctive opicapone in older adults (aged ≥65 years) using pooled data from BIPARK-1 and BIPARK-2

OBJECTIVES

- Describe results from the double-blind, placebo-controlled, pivotal Phase 3 clinical trials, BIPARK-1 (NCT01568073) and BIPARK-2 (NCT01227655)
- Demonstrate the efficacy and safety of once-daily opicapone in older patients (≥65 years) with Parkinson's disease who are experiencing "OFF" episodes
- Apply the substantial and clinically meaningful improvements found in this analysis to the real-world management of Parkinson's disease in older patients

METHODS

STUDY DESIGN

- BIPARK-1 and BIPARK-2 were similarly designed, multinational, multicenter, randomized, double-blind, placebo-controlled, parallel-group studies (**Figure 1**)
 - In BIPARK-1, participants were randomized (1:1:1:1:1) to double-blind treatment with opicapone (5, 25, or 50 mg), entacapone (200 mg), or placebo for 14–15 weeks as adjunct to their current levodopa regimen
 - In BIPARK-2, participants were randomized (1:1:1) to double-blind treatment with opicapone (25 or 50 mg) or placebo for 14–15 weeks as adjunct to their current levodopa regimen
 - In both studies, other PD medications were allowed with stable dosing regimens, and the Investigator could adjust the levodopa dose during the first 2–3 weeks according to participant response, not exceeding baseline level

Figure 1. Study Design

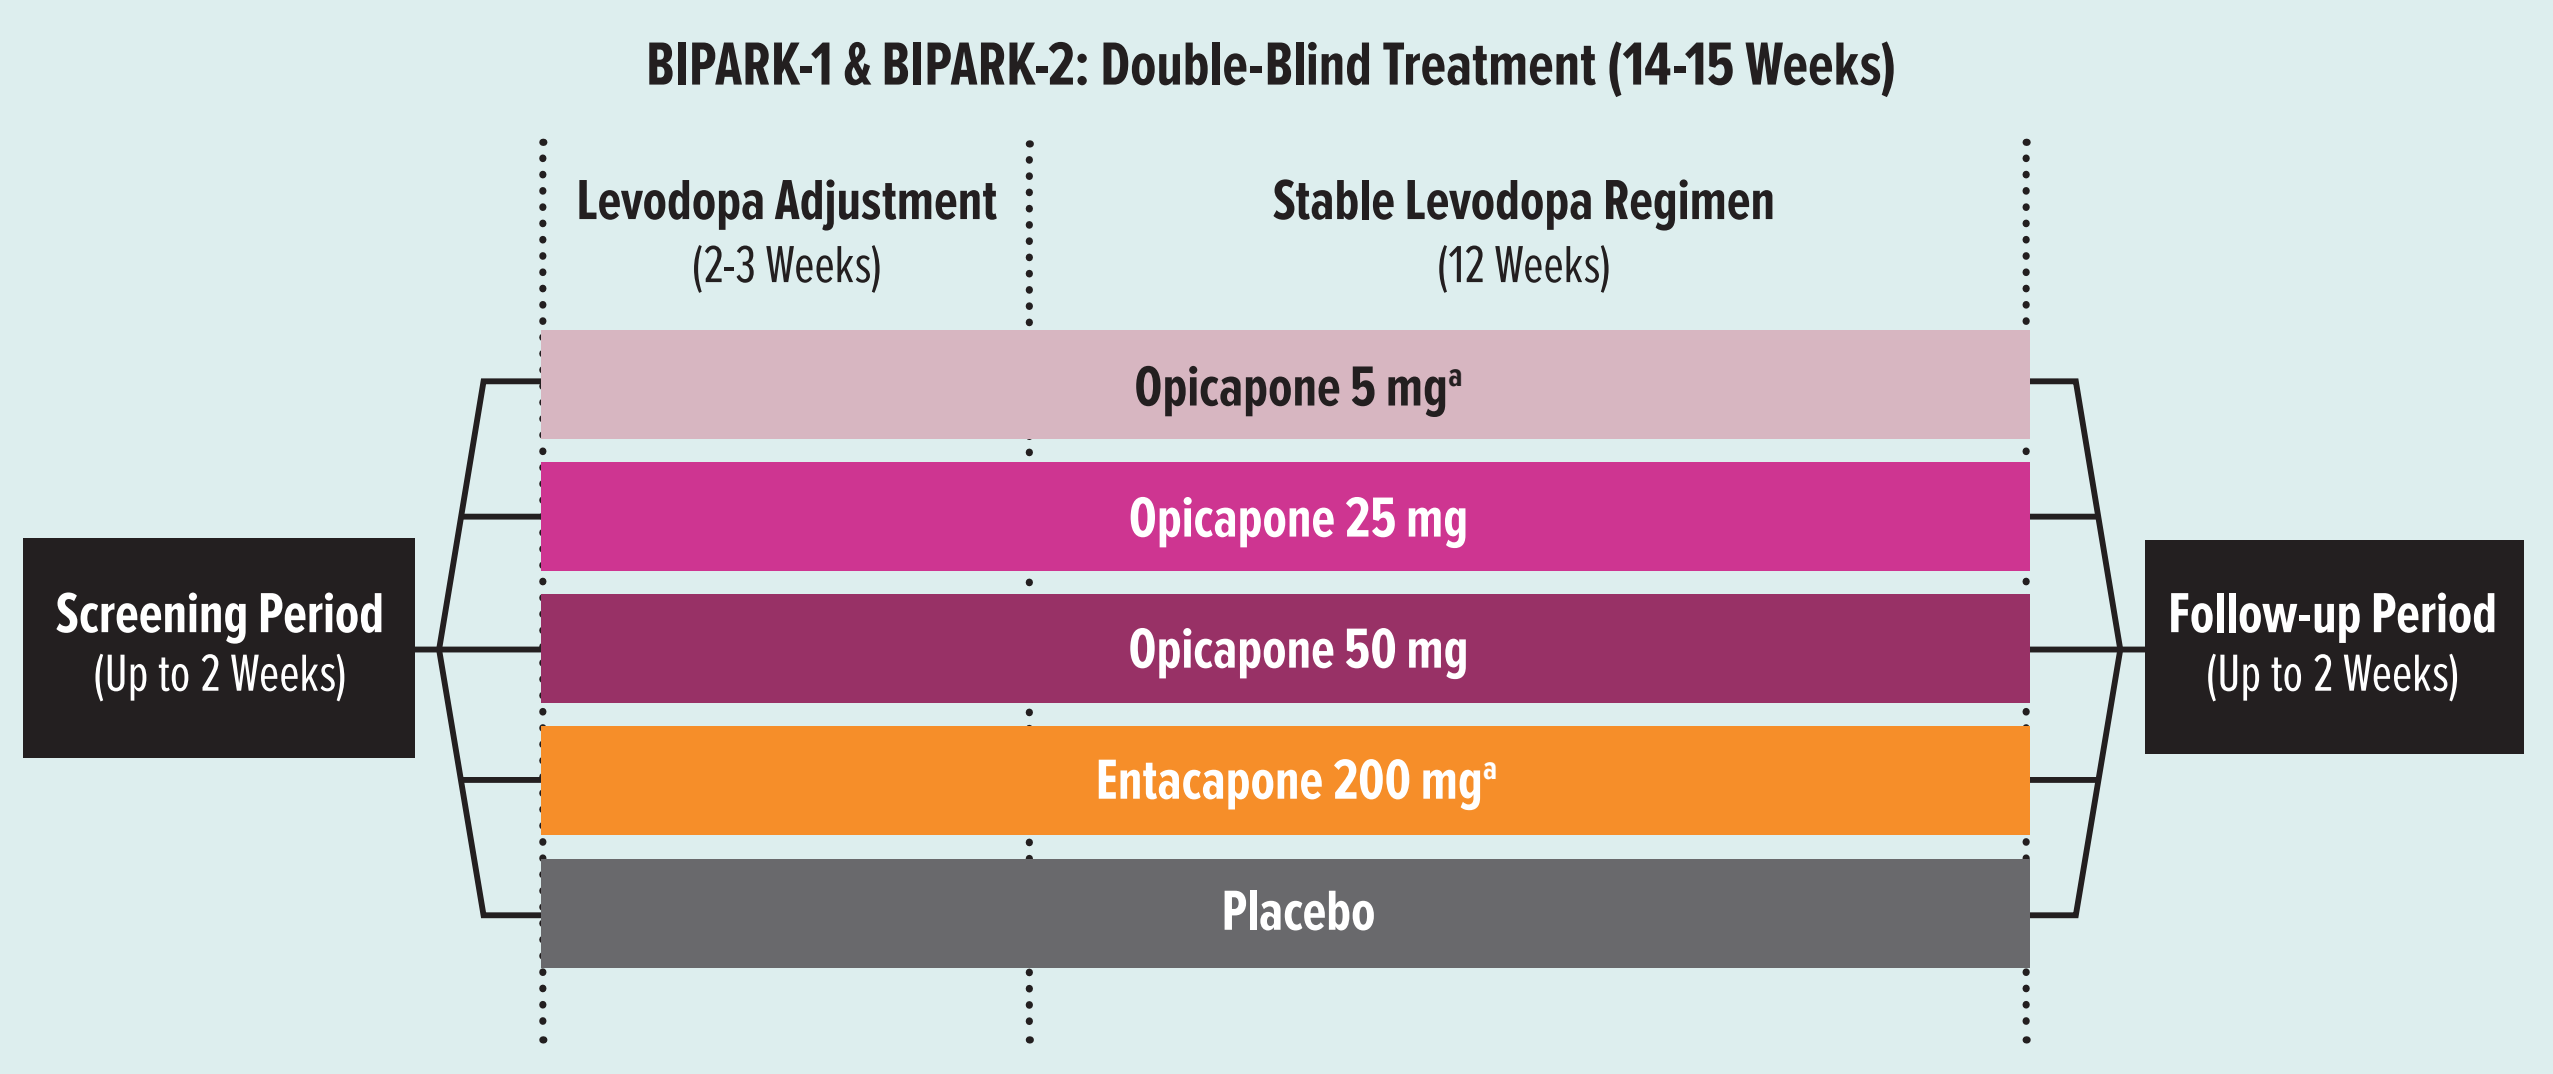
BIPARK-1 & BIPARK-2: Double-Blind Treatment (14-15 Weeks)
Levodopa Adjustment
(2-3 Weeks)
Stable Levodopa Regimen
(12 Weeks)
Screening Period
(Up to 2 Weeks)
Opicapone 5 mg[a]
Opicapone 25 mg
Opicapone 50 mg
Entacapone 200 mg[a]
Placebo
Follow-up Period
(Up to 2 Weeks)

[a]Opicapone 5 mg and entacapone 200 mg were used in BIPARK-1 only; all other groups were present in both BIPARK-1 and BIPARK-2.

PARTICIPANTS

- Key inclusion criteria:
 - Adults aged 30 to 83 years with a clinical diagnosis of idiopathic PD for ≥3 years
 - Modified Hoehn and Yahr stage of 1–3 in "ON"-state
 - ≥1 year of treatment with levodopa with clinical improvement
 - Motor fluctuations with a mean "OFF"-time per day of ≥1.5 hours (not including pre-dose morning akinesia)
 - Ability to keep accurate 24-hour diaries
- Key exclusion criteria:
 - Dyskinesia disability score >3 on the Unified Parkinson's Disease Rating Scale (item 33)
 - Severe and/or unpredictable "OFF" periods
 - Previous or planned stereotaxic surgery for PD (including deep brain stimulation)
 - Previous use of entacapone (BIPARK-1 only)

POST-HOC ANALYSES

- Efficacy data from BIPARK-1 and BIPARK-2 were pooled and analyzed using a mixed model for repeated measures; analyses evaluated both older patients (aged ≥65 years) and the overall population in the placebo group and the opicapone 50 mg group (recommended opicapone dose for patients without hepatic impairment)
- In the pooled Full Analysis Set (FAS; patients who received placebo or opicapone and had ≥1 post-baseline efficacy measurement); change from baseline in absolute "OFF"-time and change from baseline in absolute "ON"-time without troublesome dyskinesia were analyzed at Week 14/15
- In the pooled Safety Population (patients who received placebo or opicapone), assessments included treatment-emergent adverse events (TEAEs; also assessed in the opicapone 25 mg group), adverse events of special interest (AESIs), clinical laboratory tests, vital signs, and electrocardiograms (ECGs); safety data were analyzed descriptively
- Numbers needed to harm (NNHs) were calculated for TEAEs (1 divided by the difference in TEAE incidence [opicapone - placebo]), with higher NNHs indicating that more patients were needed for a TEAE with opicapone to be observed

RESULTS

- Demographics and PD characteristics were generally comparable between groups (**Table 1**)

Table 1. Demographics and Baseline Characteristics (Safety Population)

	Placebo (n=257)	Opicapone 50 mg (n=265)
Demographics		
Age, mean (SD), years	62.8 (9.1)	64.5 (8.8)
Male, n (%)	142 (55.3)	160 (60.4)
White, n (%)	211 (82.1)	231 (87.2)
PD Characteristics		
Disease duration, mean (SD), years	7.7 (3.9)	7.6 (4.3)
Motor fluctuation duration, mean (SD), years	2.6 (2.2)	2.7 (2.9)
Presence of dyskinesia, n (%)	122 (47.5)	133 (50.2)
UPDRS III in "ON"-state, mean (SD)	24.8 (12.1)	25.1 (13.2)
Daily levodopa dose, mean (SD), mg	695 (321)	698 (322)
Concurrent Medications, n (%)[a]		
Dopamine agonist	188 (73.2)	183 (69.1)
Monoamine oxidase-B inhibitor	49 (19.1)	57 (21.5)

[a]Patients were allowed to select ≥1 adjunctive medication.
FAS, Full Analysis Set; PD, Parkinson's disease; SD, standard deviation; UPDRS, Unified Parkinson's Disease Rating Scale.

EFFICACY

- In both the overall FAS and in older patients, the reduction in absolute "OFF"-time from baseline to Week 14/15 was significantly greater in the opicapone 50 mg group versus placebo (**Figure 2**)

Figure 2. Change from Baseline in Absolute "OFF"-Time at Week 14/15

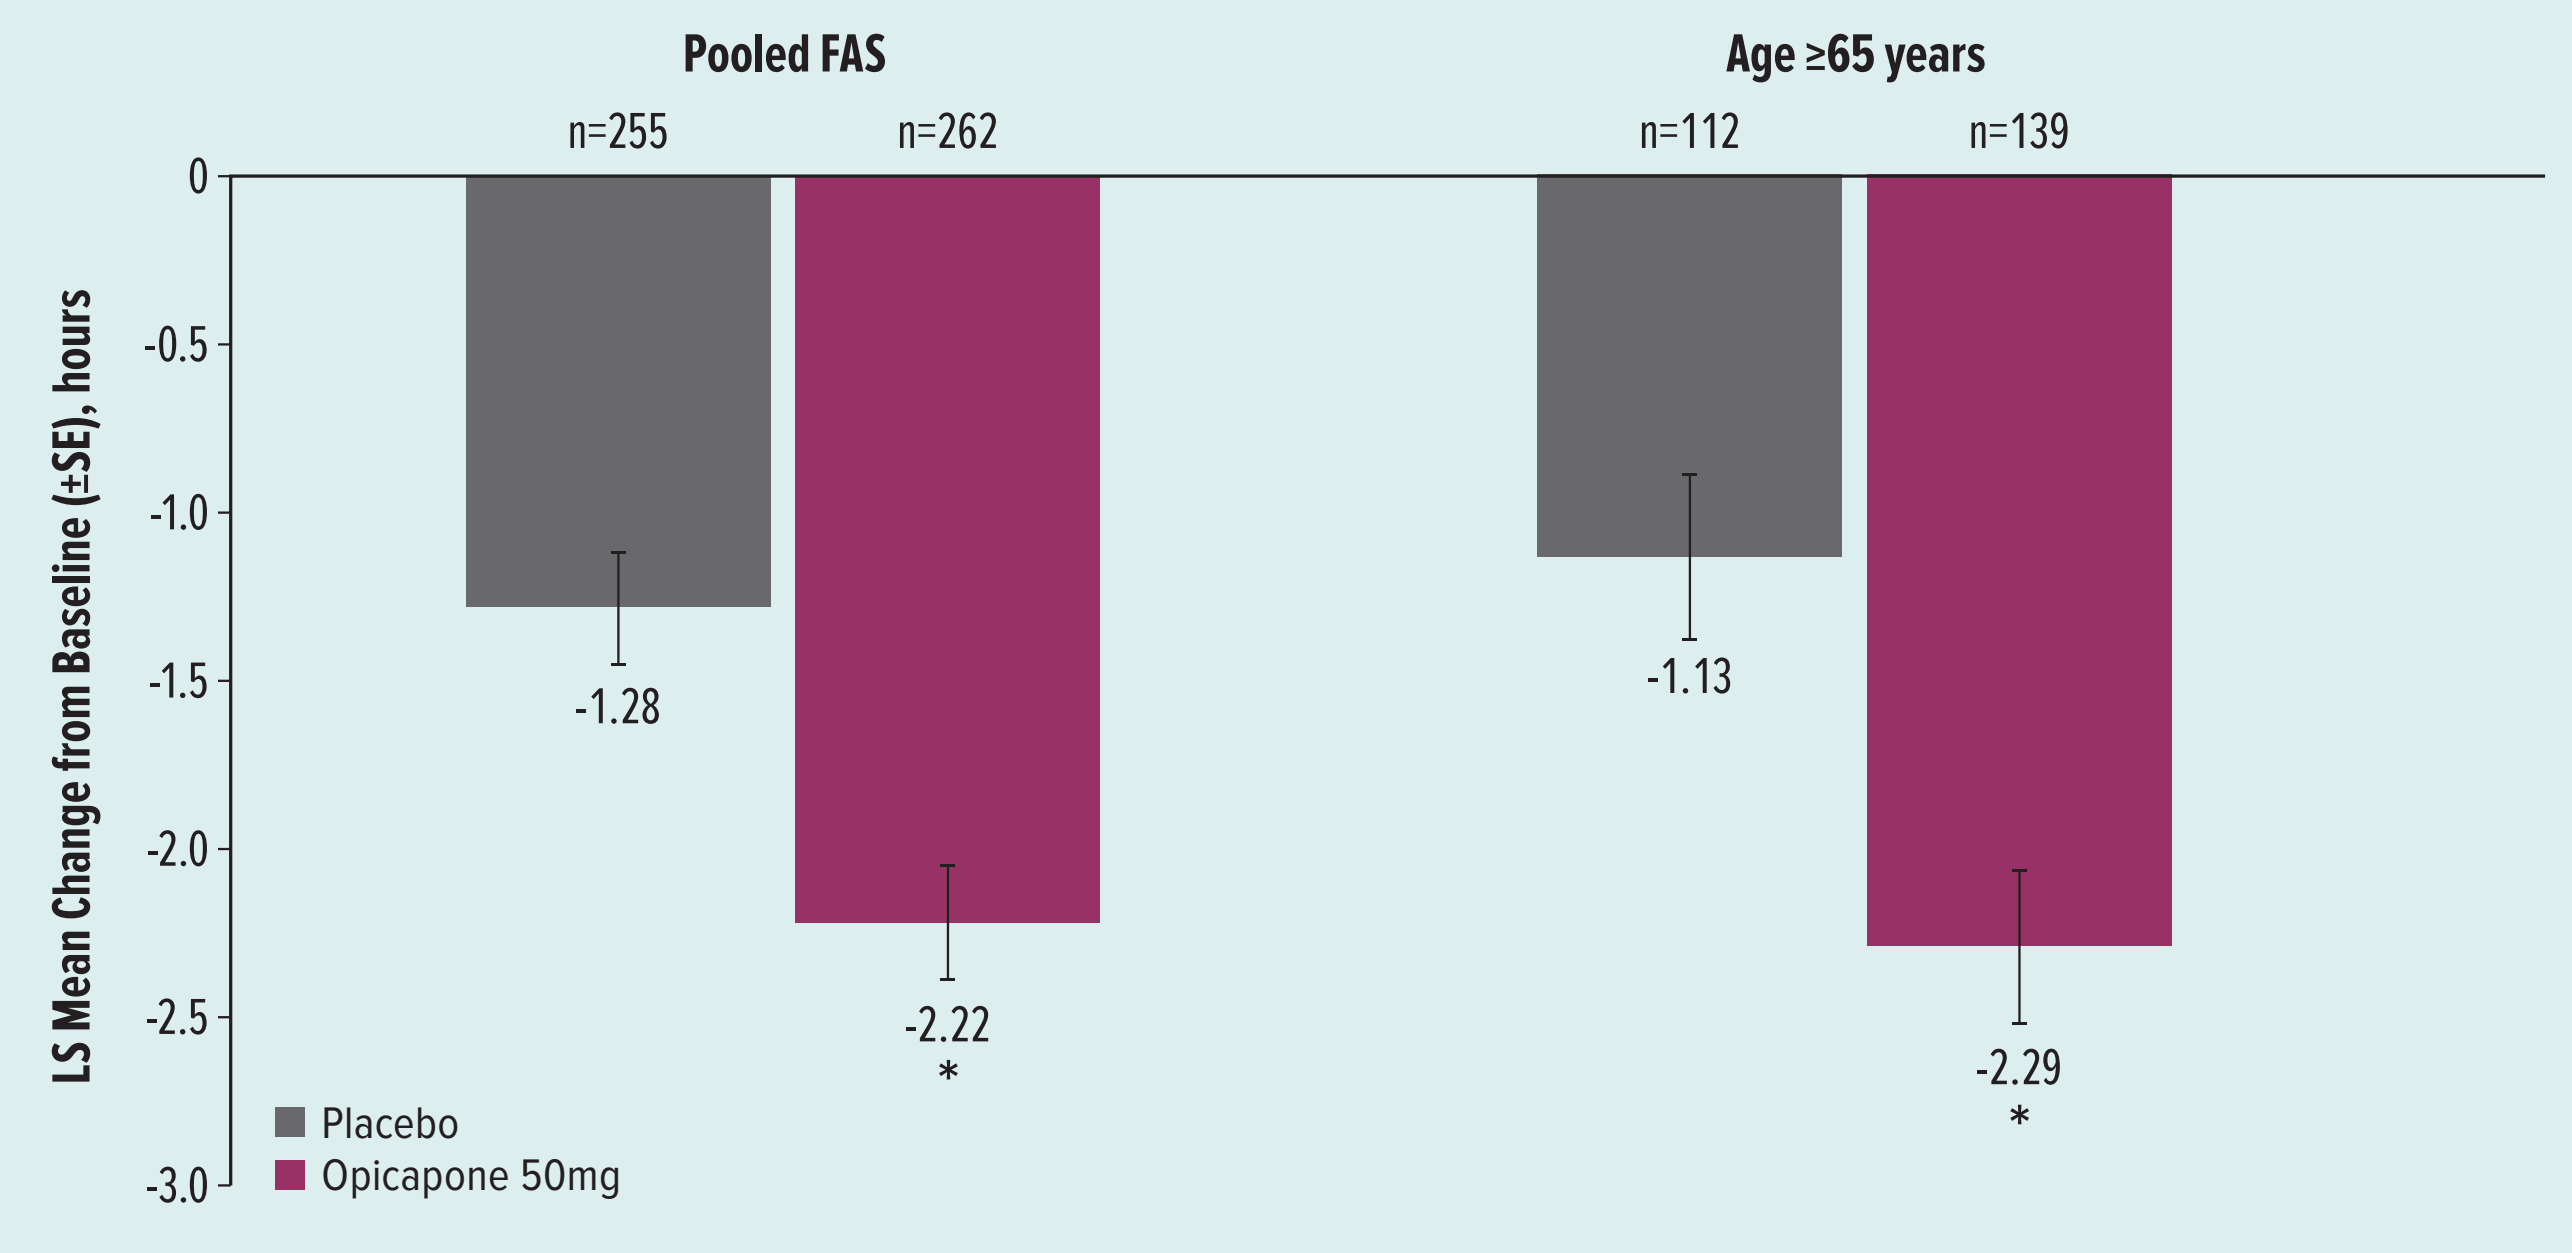
Pooled FAS
Age ≥65 years
n=255
n=262
n=112
n=139
LS Mean Change from Baseline (±SE), hours
0
-0.5
-1.0
-1.5
-2.0
-2.5
-3.0
-1.28
-2.22 *
-1.13
-2.29 *
Placebo
Opicapone 50mg

Nominal *P<0.001 versus placebo.
FAS, Full Analysis Set; LS, least squares; SE, standard error.

- In both the overall FAS and in older patients, the increase in absolute "ON"-time from baseline to Week 14/15 was significantly greater in the opicapone 50 mg group versus placebo (**Figure 3**)

Figure 3. Change from Baseline in Absolute "ON"-Time without Troublesome Dyskinesia at Week 14/15

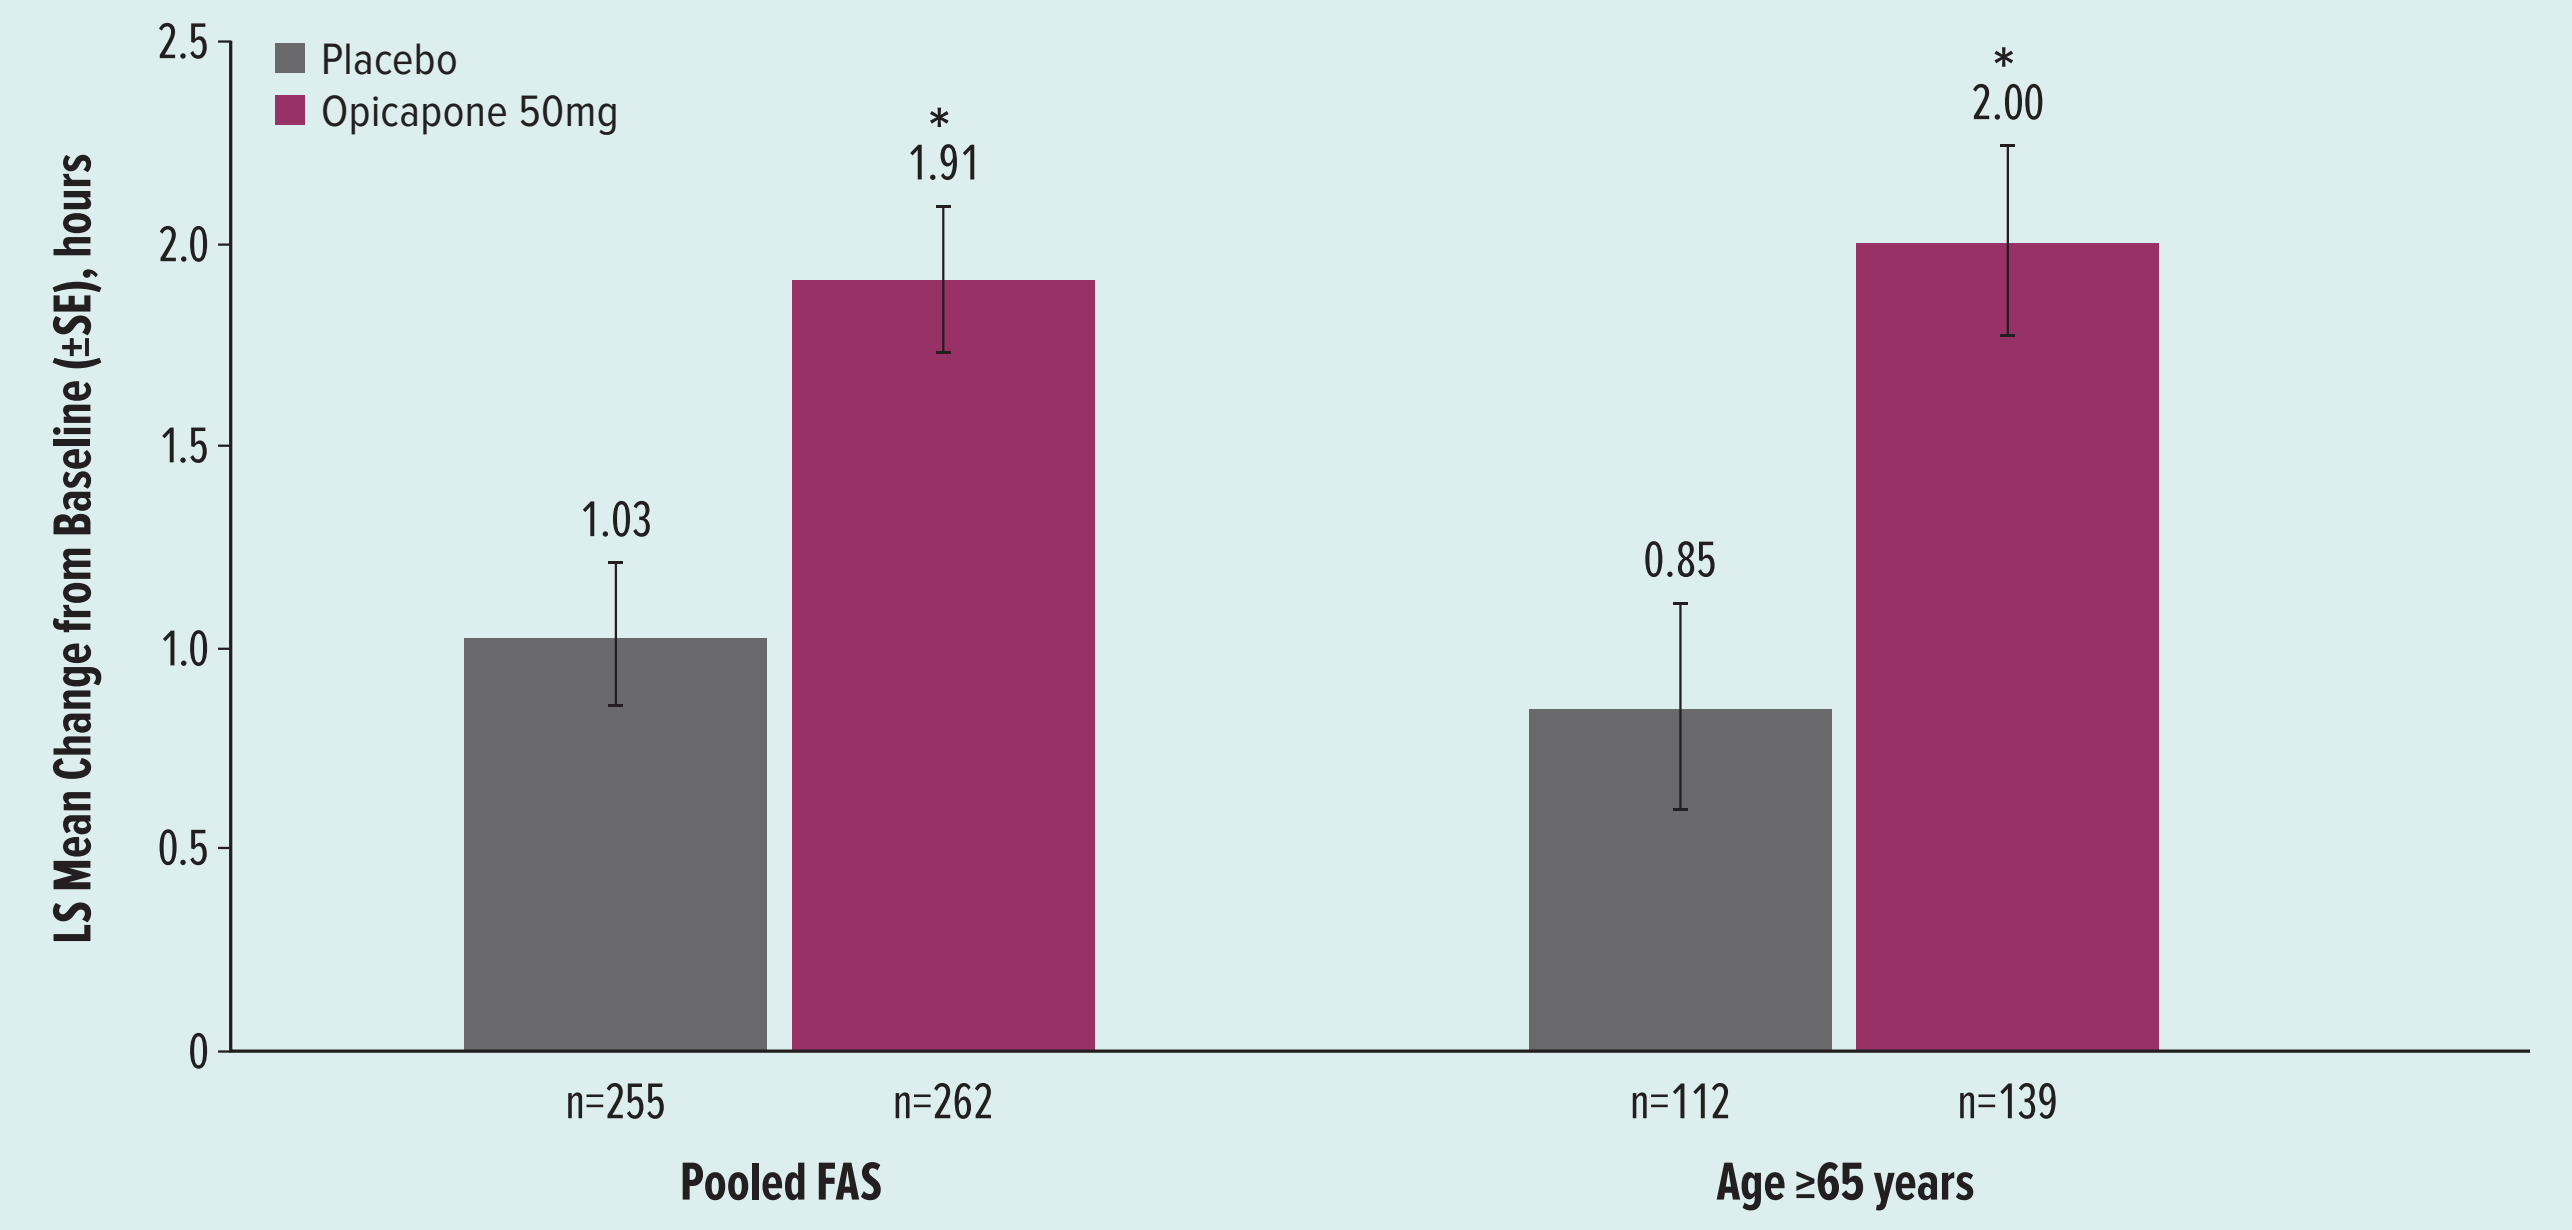
Placebo
Opicapone 50mg
LS Mean Change from Baseline (±SE), hours
2.5
2.0
1.5
1.0
0.5
0
1.03
* 1.91
0.85
* 2.00
n=255
n=262
n=112
n=139
Pooled FAS
Age ≥65 years

Nominal *P≤0.001 versus placebo.
FAS, Full Analysis Set; LS, Least Squares; SE, Standard Error.

SAFETY

- The overall incidence of TEAEs was slightly higher in older patients versus the overall pooled Safety Population in both opicapone 50 mg and placebo groups, but few patients in either group had a serious TEAE or discontinued due to a TEAE (**Table 2**)
- Opicapone 25 mg did not result in fewer TEAEs than opicapone 50 mg in either the overall Safety Population (25 mg, 62.3%; 50 mg, 64.2%) or in older patients (25 mg, 68.2%; 50 mg, 66.9%)
- The most common TEAE was dyskinesia in both the overall Safety Population and in older patients, and few patients had serious dyskinesia in either the overall Safety Population (placebo, 0%; opicapone 50 mg, 0.4%) or in older patients (placebo, 0%; opicapone 50 mg, 0.7%)
- Similarly, few patients discontinued the study due to dyskinesia in either the overall Safety Population (placebo, 0.4%; opicapone 50 mg, 3.0%) or in older patients (placebo, 0%; opicapone 50 mg, 2.2%)

Table 2. Treatment-Emergent Adverse Events

	Age ≥ 65 years			Pooled Safety Population		
	Placebo (n=112) n (%)	Opicapone 50 mg (n=139) n (%)	NNH[a]	Placebo (n=257) n (%)	Opicapone 50 mg (n=265) n (%)	NNH[a]
Summary						
Any TEAE	69 (61.6)	93 (66.9)	18	147 (57.2)	170 (64.2)	14
Any serious TEAE	8 (7.1)	8 (5.8)	-73	11 (4.3)	13 (4.9)	159
Any TEAE leading to discontinuation[b]	5 (4.5)	15 (10.8)	15	19 (7.4)	24 (9.1)	60
Deaths[c]	0 (0.0)	0 (0.0)	N/A	1 (0.4)	0 (0.0)	-257
Common TEAEs[d]						
Dyskinesia	7 (6.3)	25 (18.0)	8	16 (6.2)	54 (20.4)	7
Constipation	2 (1.8)	12 (8.6)	14	5 (1.9)	17 (6.4)	22
Insomnia	3 (2.7)	5 (3.6)	108	4 (1.6)	9 (3.4)	54
Dry mouth	3 (2.7)	5 (3.6)	108	3 (1.2)	8 (3.0)	54
Common AESIs						
Fall	8 (7.1)	5 (3.6)	-29	12 (4.7)	8 (3.0)	-61
Somnolence	2 (1.8)	1 (0.7)	-94	5 (1.9)	5 (1.9)	-1703

[a]Negative NNH indicates lower incidence for opicapone versus placebo.
[b]Includes 3 participants who had a TEAE that started during the double-blind period and resulted in discontinuation from the open-label extension period.
[c]One death in the placebo group due to pneumonia.
[d]Reported in ≥5% of participants in any treatment group.
AESI, adverse event of special interest; NNH, number needed to harm; TEAE, treatment-emergent adverse event.

- Due to the low incidence of AESIs (<5% in any treatment group), subgroup analyses for other AESIs associated with COMT inhibitors (e.g., diarrhea, hepatotoxicity) were not performed
- There were no clinically important differences or consistent trends between opicapone 50 mg and placebo groups for mean changes from baseline in clinical laboratory tests, vital signs, or ECGs

CONCLUSIONS

- In the pivotal Phase 3 studies BIPARK-1 and BIPARK-2, levodopa-treated adults with PD and motor fluctuations had a significantly greater reduction in daily "OFF"-time with adjunctive opicapone 50 mg versus placebo
- Post-hoc pooled analyses indicated treatment effects with opicapone 50 mg in patients ≥65 years old were comparable to the overall study population
- Although TEAEs were slightly more common in older patients in both the opicapone 50 mg and placebo groups, opicapone 50 mg was generally well-tolerated in older patients, supporting that opicapone requires no dose adjustment for age
- These results indicate that adding once-daily opicapone 50 mg as adjunctive treatment to levodopa/carbidopa can be both effective and safe in older patients with PD and "OFF" episodes

REFERENCES

1. Tambasco N, Romoli M, Calabresi P. Levodopa in Parkinson's Disease: Current Status and Future Developments. *Curr Neuropharmacol.* 2018;16(8):1239-1252.
2. Lexicomp. Opicapone: Drug information. *UpToDate* 2021; https://www.uptodate.com/contents/opicapone-drug-information.
3. Ferreira JJ, Lees A, Rocha J-F, Poewe W, Rascol O, Soares-da-Silva P. Opicapone as an adjunct to levodopa in patients with Parkinson's disease and end-of-dose motor fluctuations: a randomised, double-blind, controlled trial. *The Lancet Neurology.* 2016;15(2):154-165.
4. Lees AJ, Ferreira J, Rascol O, et al. Opicapone as Adjunct to Levodopa Therapy in Patients With Parkinson Disease and Motor Fluctuations: A Randomized Clinical Trial. *JAMA Neurol.* 2017;74(2):197-206.

DISCLOSURES: This study was supported by BIAL – Portela & C[a] S.A., Coronado, Portugal. This poster was funded by Neurocrine Biosciences, Inc., San Diego, CA. Writing assistance and editorial support were provided by Prescott Medical Communications Group, Chicago, IL. Please email medinfo@neurocrine.com if you have any questions on this presentation.

PRESENTED AT THE NEUROSCIENCE ADVANCED PRACTICE PROVIDER EDUCATIONAL CONFERENCE
JANUARY 5-8, 2022; SAN DIEGO, CA