For additional information, please contact Neurelis Medical Information at medinfo@neurelis.com

Evaluation of Diazepam Nasal Spray in Patients With Seizure Clusters Concomitantly Receiving Clobazam: A Subgroup Analysis From a Completed Phase 3, Long-Term, Open-Label Safety Study

Michael R. Sperling, MD[1]; Daniel Tarquinio, DO[2]; James W. Wheless, MD[3]; Gregory D. Cascino, MD[4]; Jay Desai, MD[5]; R. Edward Hogan, MD[6]; Kore Liow, MD[7]; David F. Cook, PhD[8]; Adrian L. Rabinowicz, MD[8]; Enrique Carrazana, MD[8]

[1]Thomas Jefferson University, Philadelphia, PA; [2]Center for Rare Neurological Diseases, Atlanta, GA; [3]Le Bonheur Children's Hospital, University of Tennessee Health Science Center, Memphis, TN; [4]Mayo Clinic, Rochester, MN; [5]Children's Hospital of Los Angeles, Los Angeles, CA; [6]Washington University in St. Louis, St. Louis, MO; [7]Hawaii Pacific Neuroscience, Honolulu, HI; [8]Neurelis, Inc., San Diego, CA

Background

- Many patients with epilepsy take a regimen of antiseizure medications (ASMs) that include maintenance benzodiazepines[1]
 - For example, clobazam is indicated for seizures associated with Lennox-Gastaut syndrome, a severe form of epilepsy that is refractory to most ASMs[1,2]
- Benzodiazepines are the mainstay of rescue therapy for seizure clusters[3]
- Diazepam nasal spray (Valtoco®), is approved by the US Food and Drug Administration (FDA) for the acute treatment of intermittent, stereotypic episodes of frequent seizure activity (ie, seizure clusters, acute repetitive seizures) that are distinct from a patient's usual seizure pattern in patients with epilepsy aged 6 years and older[4]
 - The intranasal route of administration for acute rescue therapy has the advantages of being noninvasive, being easy to access, allowing rapid onset of effect, avoiding first-pass metabolism, and being socially acceptable compared with rectal administration[5]
 - The FDA states that diazepam nasal spray improves ease of use because it is less invasive and easier to administer during a seizure, compared with rectal administration[6]
- Whether use of chronic benzodiazepines, such as clobazam, influences the effectiveness of diazepam nasal spray as rescue medication for seizure clusters is uncertain and of importance

Objective

- To evaluate the long-term effectiveness and safety of diazepam nasal spray as rescue medication for treatment of seizure clusters in subgroups of patients receiving clobazam, which is used chronically, or other benzodiazepines, which are typically used intermittently in epilepsy, as part of their ASM regimen in a long-term safety study

Methods

- The results presented here are from the final analysis of a phase 3, open-label, repeat-dose safety study of diazepam nasal spray (ClinicalTrials.gov identifier: NCT02721069) that was completed in July 2020[7]
 - Received institutional review board approval
 - Conducted in accordance with the Declaration of Helsinki
 - Written informed consent was obtained from all patients
- Enrolled patients had a clinical diagnosis of epilepsy and, in the opinion of the investigator, might need benzodiazepine treatment for seizure control once every other month on average (ie, 6 times a year) despite a stable regimen of ASMs
- Key inclusion criteria
 - Male or female patients aged 6–65 years
 - Diagnosis of partial or generalized epilepsy with motor seizures or seizures with clear alteration of awareness
 - Availability of a qualified caregiver or medical professional who could administer study medication in the event of a seizure
 - No clinically significant abnormal findings in the patient's medical history, or on physical examination, electrocardiogram, or clinical laboratory results during screening
 - Female patients of childbearing potential agreed to use an approved method of birth control
- History of status epilepticus or seasonal allergies/rhinitis was permitted; no restriction was made on concomitant use of benzodiazepines
- Key exclusion criteria
 - History of a clinically significant medical condition that would jeopardize the safety of the patient
 - Major depression or a past suicide attempt or suicidal ideation
- Patients and care partners were trained on the proper use of the nasal sprayer device at screening and as needed during the study
- Diazepam nasal spray was administered in doses of 5, 10, 15, or 20 mg, based on patient age and weight
 - If needed, a second dose could be administered 4–12 hours after the first dose
 - Investigators could adjust doses for effectiveness or safety if there were no safety concerns associated with the change

Subgroup Analysis

- Seizure timing and study drug administration were recorded in patient diaries and reported at regularly scheduled visits; treatment-emergent adverse events (TEAEs) were collected
- Second doses were assessed during the 24 hours after the initial dose
- The proportion of seizure events treated with a second dose of diazepam nasal spray was used as an exploratory, proxy measure for effectiveness
- Subgroup analysis was performed to compare those taking clobazam with those taking other benzodiazepines
- Safety measures, including TEAEs and their possible relationship to the study drug, were assessed
 - TEAEs were summarized, and descriptive statistics were calculatedw

Results

Patients Overall

- A total of 175 patients were enrolled in this study; 12 patients discontinued before dosing, and 163 patients (54% female; mean age, 23.1 years) received ≥1 dose of diazepam nasal spray and were included in the safety population
- The majority of these patients (81.6%) had a duration of exposure to diazepam nasal spray of ≥12 months, and there was a mean of 2.3 doses per month
- The 163 patients in the safety population experienced a total of 3853 seizure clusters and 4390 doses of diazepam nasal spray were administered
 - Of the 3853 seizure clusters, 485 (12.6%) required a second dose of diazepam nasal spray within 24 hours of the initial dose

Patients Taking Concomitant Benzodiazepines

- Among the 163 patients, 125 (76.7%) took concomitant benzodiazepines and are included in the current analysis
- Forty-six (36.8%) of the 125 patients used clobazam and 79 (63.2%) used other benzodiazepines (**Table 1**)
 - The mean age of the clobazam patients (17.7 years) was lower than in the other benzodiazepines subgroup (26.0 years)
 - Exposure to diazepam nasal spray was ≥12 months for most patients (clobazam subgroup, 91.3%; other benzodiazepines subgroup, 81.0%)

Table 1. Baseline Characteristics and Diazepam Nasal Spray Exposure and Dosing

Variable	Clobazam Subgroup (n=46)	Other Benzodiazepines Subgroup* (n=79)
Sex, n (%)		
Male	20 (43.5)	39 (49.4)
Female	26 (56.5)	40 (50.6)
Age, y		
Mean (SD)	17.7 (11.8)	26.0 (16.0)
Median (range)	13.0 (6–48)	24.0 (6–65)
Race, n (%)		
White	38 (82.6)	67 (84.8)
Black or African American	5 (10.9)	7 (8.9)
Asian	1 (2.2)	2 (2.5)
Other	2 (4.3)	3 (3.8)
Duration of exposure, months, n (%)		
<6	1 (2.2)	5 (6.3)
6–<12	3 (6.5)	10 (12.7)
≥12	42 (91.3)	64 (81.0)
Seizure clusters treated	1201	2198
Seizure clusters with second dose administered	120	306

*Clonazepam (n=54), diazepam (n=50), lorazepam (n=37), midazolam (n=9); clorazepate dipotassium (n=6); alprazolam (n=5).

- Retention rates (study completion irrespective of study closure) were 80.4% in the clobazam subgroup and 72.2% in the other benzodiazepines subgroup
- The subgroups had similar mean total doses of diazepam nasal spray per patient (clobazam subgroup, 26.1; other benzodiazepines subgroup, 27.8) and doses per month (clobazam subgroup, 2.5; other benzodiazepines subgroup, 2.3)
- In both subgroups, <15% of seizure episodes were treated with a second dose (**Figure 1**)

Figure 1. Seizure Episodes Treated with a Second Dose of Diazepam Nasal Spray

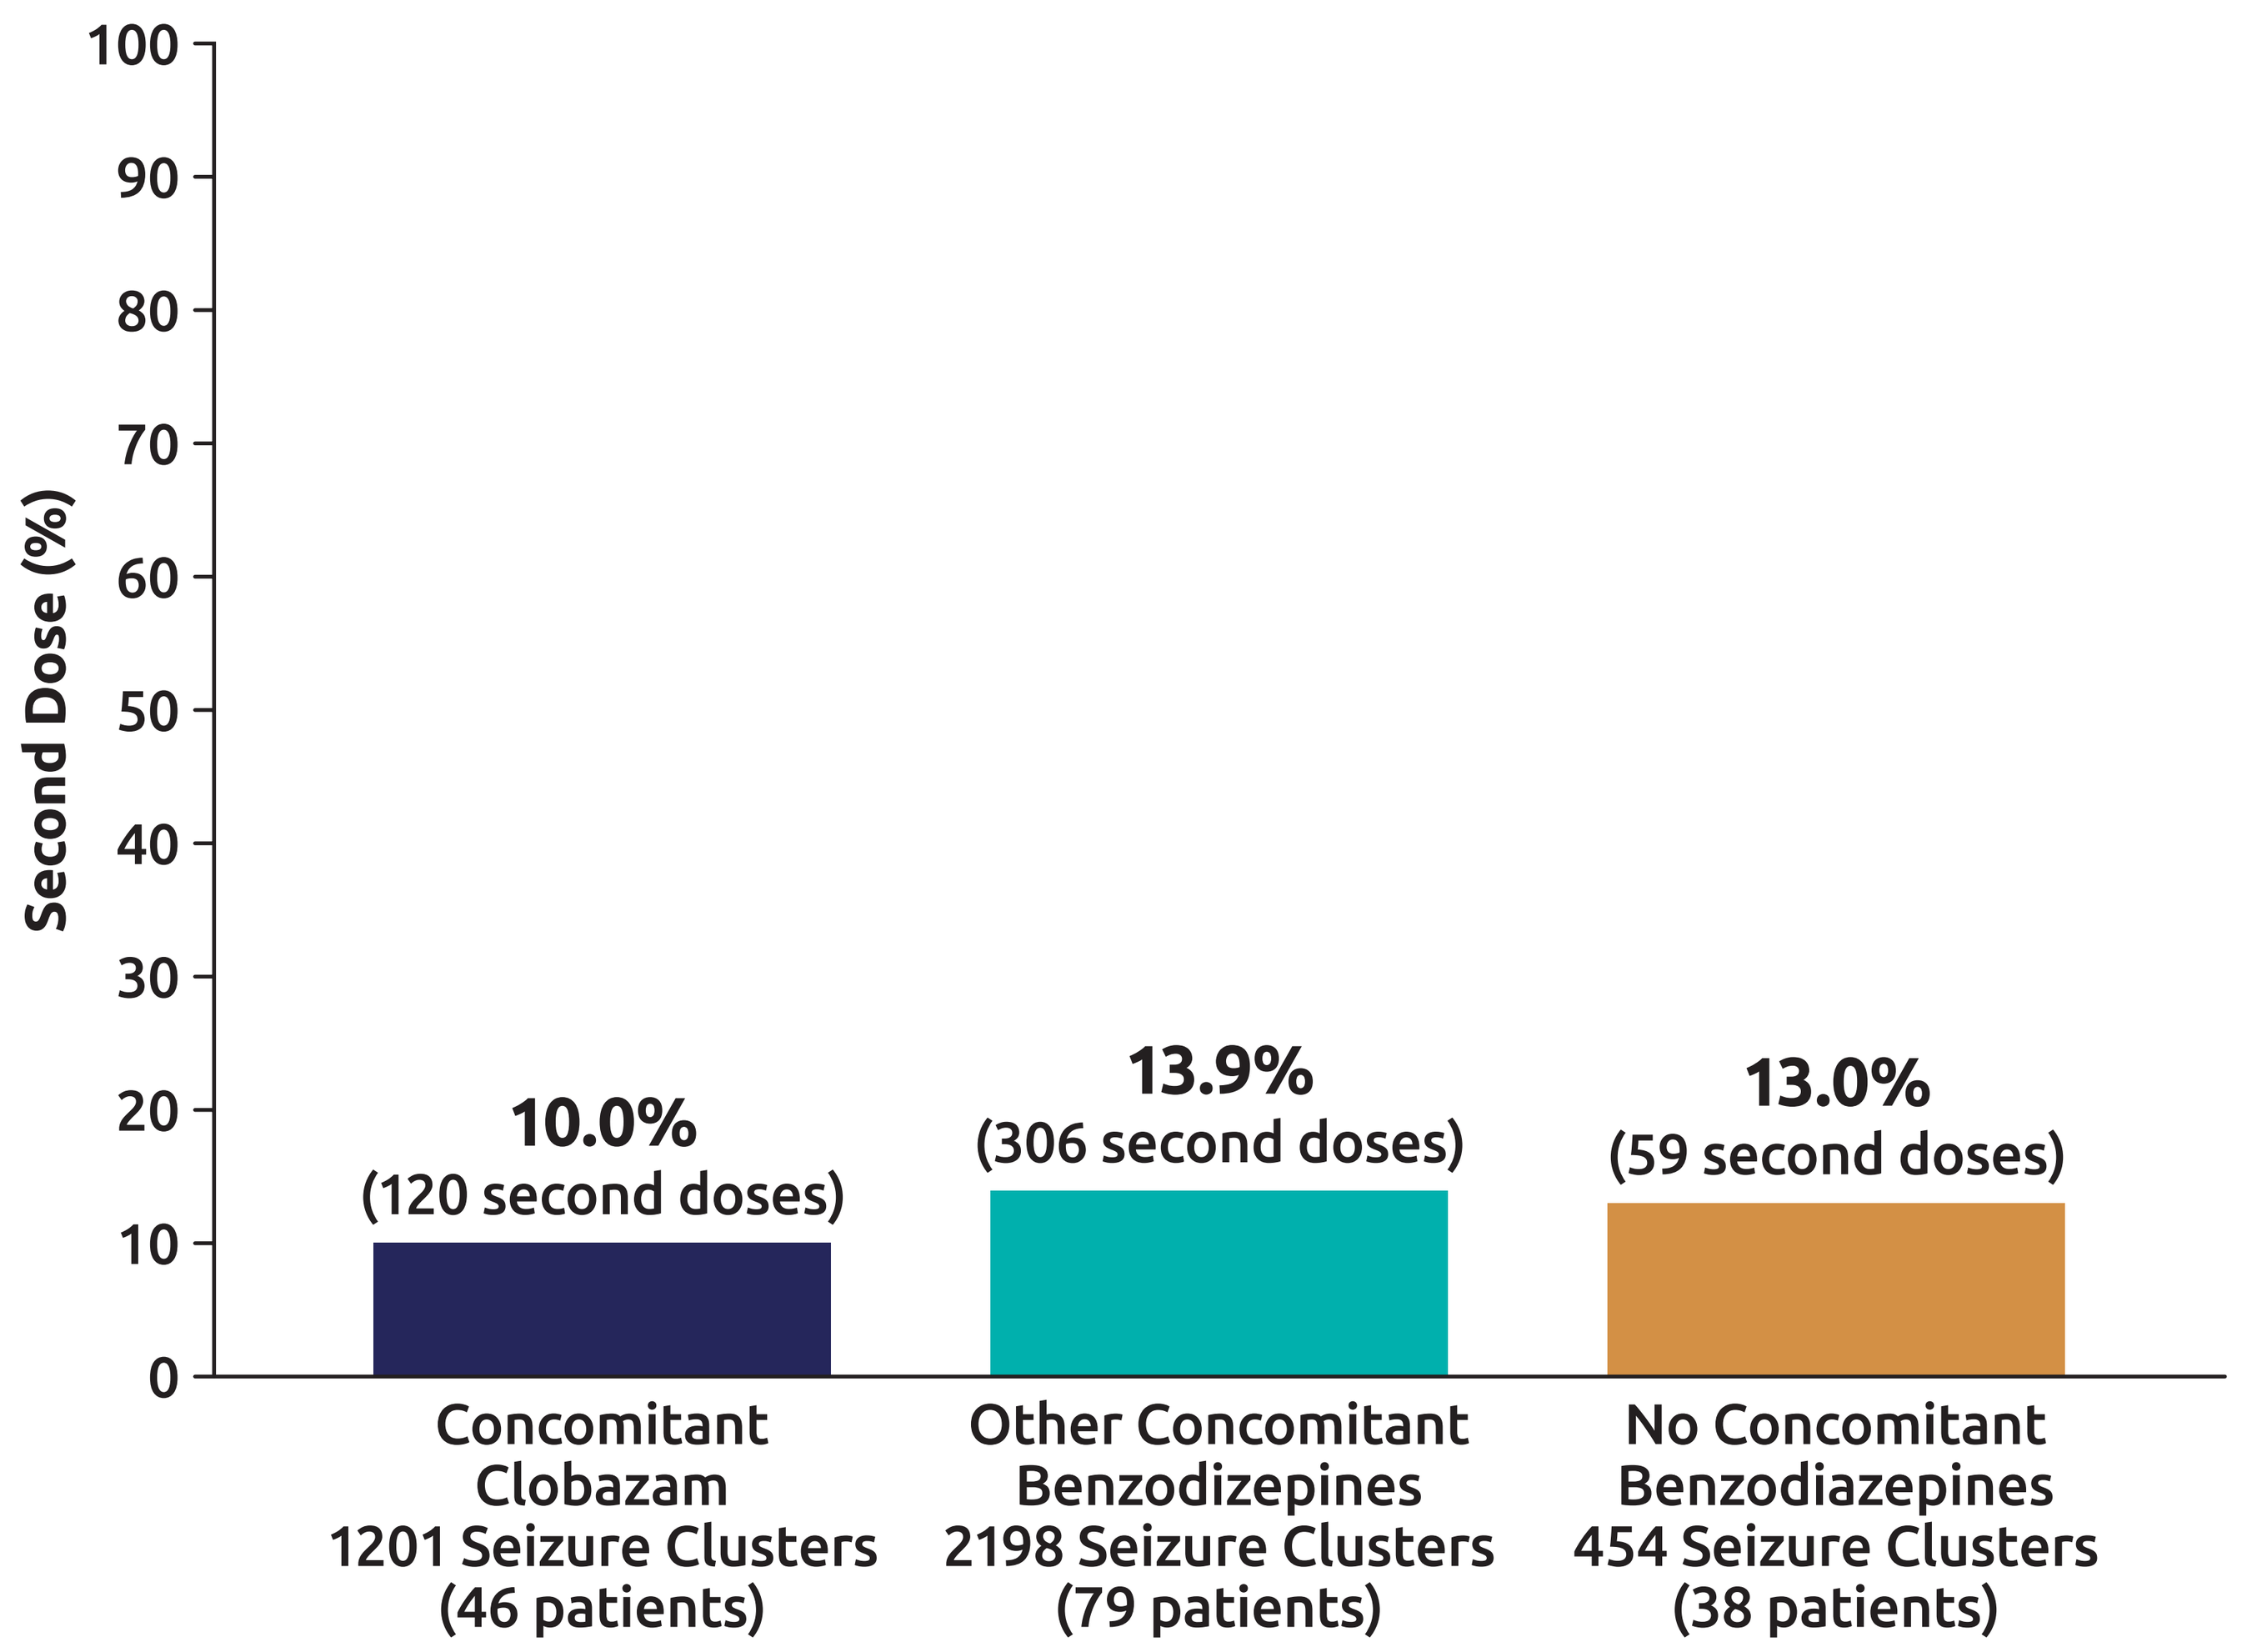
Second Dose (%)
100
90
80
70
60
50
40
30
20
10
0
10.0%
(120 second doses)
13.9%
(306 second doses)
13.0%
(59 second doses)
Concomitant Clobazam
1201 Seizure Clusters
(46 patients)
Other Concomitant Benzodizepines
2198 Seizure Clusters
(79 patients)
No Concomitant Benzodiazepines
454 Seizure Clusters
(38 patients)

Safety

- The proportion of patients with TEAEs was slightly higher in the clobazam subgroup (89.1%) than in the other benzodiazepines subgroup (83.5%) (**Table 2**)
 - Rates of treatment-related TEAEs were similar (approximately 20%) in the 2 subgroups
 - Serious TEAE rates were higher among the clobazam patients (47.8% vs 25.3%); none of the serious TEAEs were considered treatment related

Table 2. TEAEs in Patients Treated With Diazepam Nasal Spray

TEAE	Clobazam Subgroup (n=46)	Other Benzodiazepines Subgroup (n=79)
Any TEAE	41 (89.1)	66 (83.5)
Serious TEAE	22 (47.8)	20 (25.3)
Treatment-related TEAE	9 (19.6)	17 (21.5)
Discontinuation due to TEAE	0	1 (1.3)[a]
Death	0	1 (1.3)[a]
Most common TEAEs (≥10% in either subgroup)		
Seizure	13 (28.3)	13 (16.5)
Nasopharyngitis	11 (23.9)	3 (3.8)
Upper respiratory tract infection	10 (21.7)	7 (8.9)
Pneumonia	7 (15.2)	4 (5.1)
Pyrexia	7 (15.2)	8 (10.1)
Urinary tract infection	7 (15.2)	3 (3.8)
Influenza	6 (13.0)	6 (7.6)
Most common treatment-related TEAEs (≥2 patients in either subgroup)		
Nasal discomfort	2 (4.3)	7 (8.9)
Dysgeusia	0	3 (3.8)
Headache	0	3 (3.8)
Migraine	0	2 (2.5)
Rhinalgia	0	2 (2.5)
Somnolence	0	2 (2.5)

TEAE, treatment-emergent adverse event.
[a]Not deemed treatment related by the investigator.

- The most common TEAEs (≥10%) generally had a higher incidence in the clobazam subgroup than in the other benzodiazepines subgroup
 - TEAEs reported in ≥20% of patients in either the clobazam subgroup or the other benzodiazepines subgroup included seizure (28.3% vs 16.5%), nasopharyngitis (23.9% vs 3.8%), and upper respiratory tract infection (21.7% vs 8.9%)
 - No events of respiratory depression were reported
 - The only treatment-related TEAE in ≥5% of patients in either group was nasal discomfort (8.9%, other benzodiazepines subgroup [4.3%, clobazam subgroup])
 - There were no discontinuations due to a treatment-related TEAE in either group

Conclusions

- The final results of this subgroup analysis show that the long-term effectiveness and safety profile of diazepam nasal spray was not substantially different between patients receiving concomitant clobazam compared with patients receiving other concomitant benzodiazepines
 - The number of second doses needed per episode (used as a proxy of effectiveness) was not affected by concomitant clobazam use
 - Furthermore, the use of clobazam did not have any clinically relevant effects on the safety/tolerability profile of diazepam nasal spray or on study retention compared with other chronic or intermittent benzodiazepines
 - These results mirror those of the overall study, which included 38 patients without concomitant benzodiazepine use
 - Please see Poster 3.274, ***Safety and Time to Second Doses in Age Subgroups of Patients With Seizure Clusters Treated With Diazepam Nasal Spray in a Phase 3, Open-Label, Repeat-Dose Safety Study***
- Overall, results from this long-term study suggest that diazepam nasal spray can be used in patients concomitantly using clobazam as part of their ASM regimen

References **1.** Segal EB, et al. *Epilepsia.* 2021;62(6):1442-1450. **2.** Chin RFM, et al. *Seizure.* 2021;91:159-166. **3.** Haut SR. *Curr Opin Neurol.* 2015;28(2):143-150. **4.** Neurelis, Inc. Valtoco (diazepam nasal spray). Full Prescribing Information. Neurelis, Inc.; 2021. **5.** Cloyd J, et al. *Epilepsia.* 2021;62(4):846-856. **6.** US Food and Drug Administration. Clinical superiority findings. Accessed June 23, 2020. https://www.fda.gov/industry/designating-orphan-product-drugs-and-biological-products/clinical-superiority-findings **7.** Wheless JW, et al. *Epilepsia.* 2021;62(10):2485-2495.

Acknowledgments Medical writing support was provided by Jordanna Bermack, PhD, for The Curry Rockefeller Group, LLC (Tarrytown, NY), and was funded by Neurelis, Inc. (San Diego, CA).

Disclosures **Dr Sperling** has received compensation for speaking at CME programs from Medscape, Projects for Knowledge, International Medical Press, Eisai, and UCB Pharma. He is an advisor for scientific publications for Neurelis, Inc. He consults for Medtronic with payments to Thomas Jefferson University. He has received research support from Eisai Inc.; Medtronic; Neurelis, Inc.; SK Life Science; Takeda; Xenon; Cerevel; UCB Pharma; and Engage Pharmaceuticals. He has received royalties from Oxford University Press. **Dr Tarquinio** has received personal compensation for consulting, serving on a scientific advisory board, speaking, or other activities with Avexis; Marinus; and Neurelis, Inc. **Dr Wheless** has served as an advisor or consultant for CombiMatrix; Eisai Inc.; GW Pharmaceuticals; Lundbeck, Inc.; Neurelis, Inc; NeuroPace, Inc.; Supernus Pharmaceuticals, Inc.; and Upsher-Smith Laboratories, Inc. Dr Wheless has served as a speaker or a member of a speakers bureau for Cyberonics, Inc.; Eisai Inc.; Lundbeck, Inc.; Mallinckrodt; Neurelis, Inc,; Supernus Pharmaceuticals, Inc.; and Upsher-Smith Laboratories, Inc., and has received grants for clinical research from Acorda Therapeutics; GW Pharmaceuticals; Insys Therapeutics, Inc.; Lundbeck, Inc.; Mallinckrodt; Neurelis, Inc.; NeuroPace, Inc.; Upsher-Smith Laboratories, Inc.; and Zogenix, Inc. **Dr Cascino** has nothing to disclose. **Dr Desai** has received research funding from Neurelis, Inc.; Novartis; Ovid; Aquestive; and UCB Pharmaceuticals. **Dr Hogan** has received research support from UCB Pharmaceuticals; Neurelis, Inc; and Biogen Inc, and is an advisor for Neurelis, Inc. **Dr Liow** has received research support from Intracellular Therapies, SK Life Science, Genentech, Biotie Therapies, Monosol, Aquestive Therapeutics, Engage Therapeutics, Xenon, Lundbeck, Biogen, Eli Lilly, Pfizer, Novartis, Sunovion, Acorda, Eisai, UCB, Livanova, Axsome, and Acadia. **Dr Cook** is an employee of and has received stock options from Neurelis, Inc. **Dr Rabinowicz** is an employee of and has received stock options from Neurelis, Inc. **Dr Carrazana** is an employee of and has received stock and stock options from Neurelis, Inc.

Neuroscience Advanced Practice Provider Educational Conference (Neuro-APP) • January 5–8, 2022 • San Diego, CA and Virtually

This research study was supported by Neurelis, Inc.

© Copyright Neurelis, Inc. All Rights Reserved. MRC-21-00136